Discovery of BTX-9341, a bifunctional degrader of CDK4 and CDK6 for HR+/HER2- breast cancer

Hannah Majeski, Akinori Okano, Kirti Chahal, Angela Pasis, Casey Carlson, Arvind Shakya, Qiao Liu, Shenlin Huang, Aparajita Hoskote Chourasia and Leah Fung

Biotheryx, Inc., San Diego, CA

San Antonio Breast Cancer Symposium – December 5-9, 2023

BACKGROUND

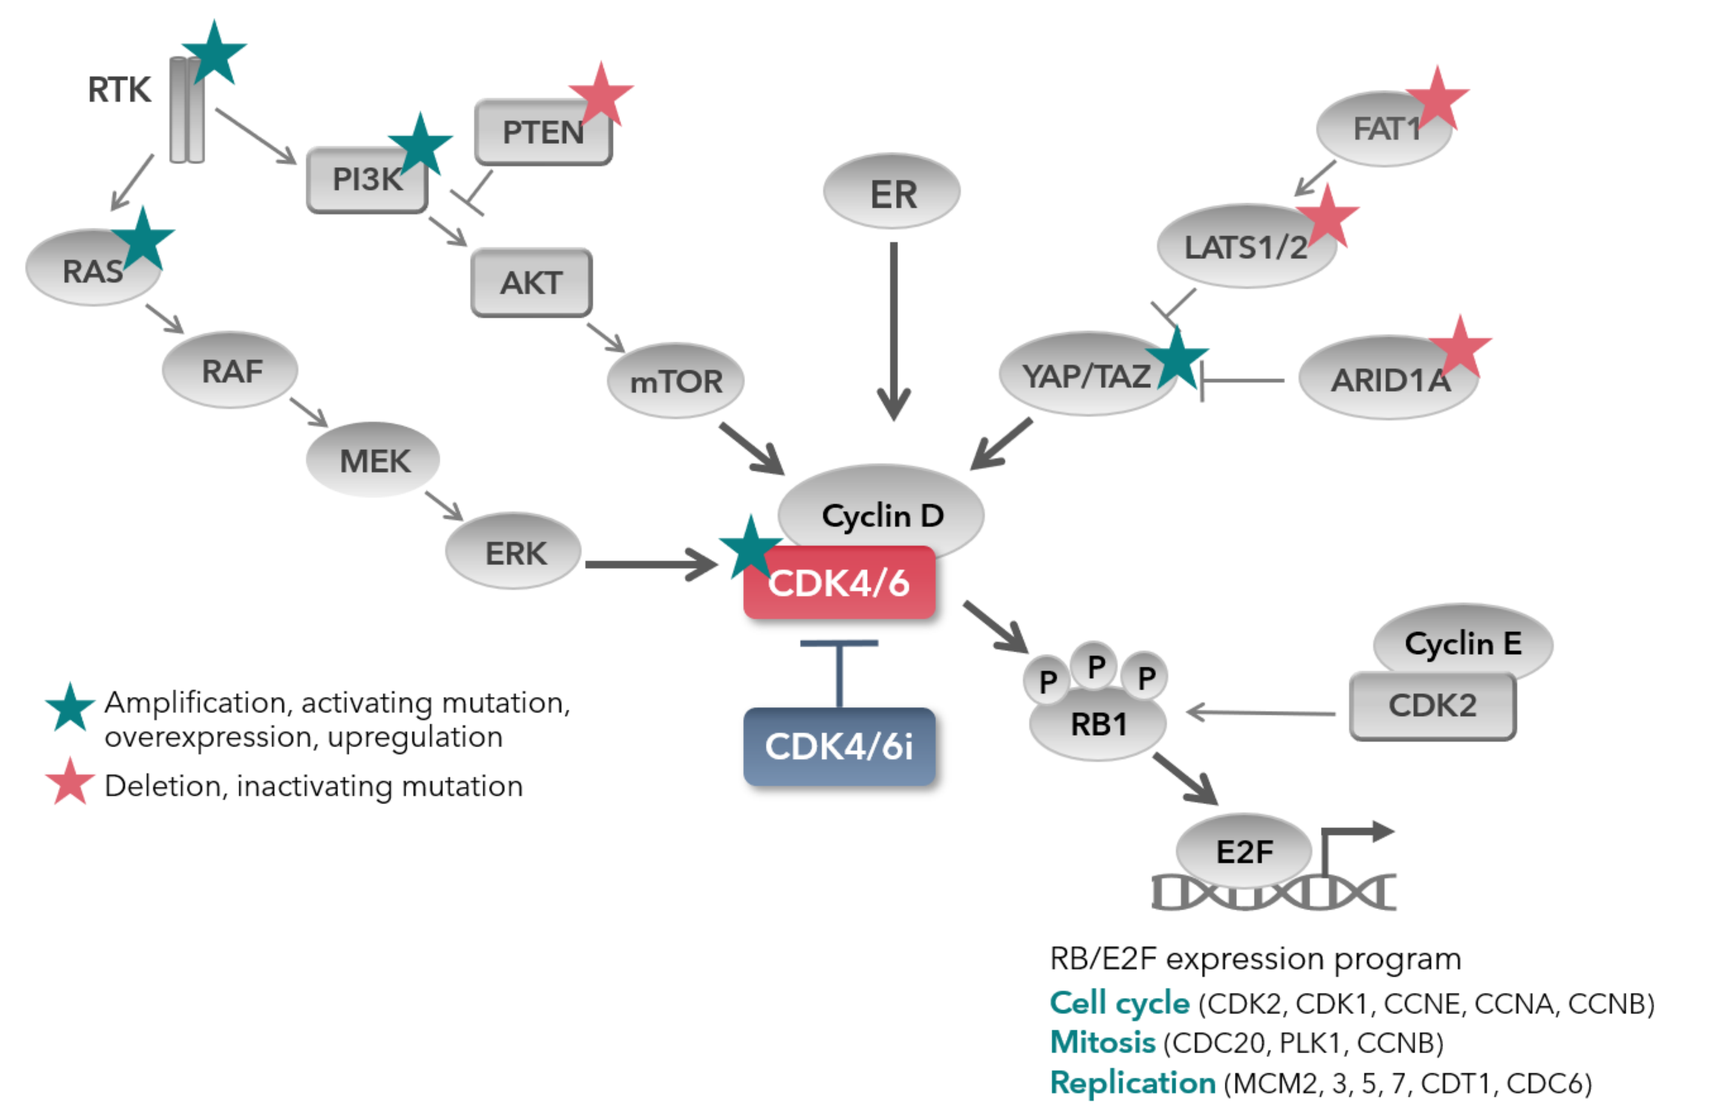
RTK
PI3K
PTEN
RAS
AKT
RAF
mTOR
MEK
ERK
ER
FAT1
LATS1/2
YAP/TAZ
ARID1A
Cyclin D
CDK4/6
CDK4/6i
Cyclin E
CDK2
RB1
E2F
Amplification, activating mutation, overexpression, upregulation
Deletion, inactivating mutation
RB/E2F expression program
Cell cycle (CDK2, CDK1, CCNE, CCNA, CCNB)
Mitosis (CDC20, PLK1, CCNB)
Replication (MCM2, 3, 5, 7, CDT1, CDC6)

CDK4 and CDK6 are kinases which regulate cell cycle progression through the phosphorylation of retinoblastoma protein (RB) which releases the transcription factor E2F, driving the expression of cell cycle promoting genes. CDK4/6 are clinically validated targets in ER+/HER2- breast cancer, with multiple CDK4/6 inhibitors (CDK4/6i) approved for use in this indication, but resistance remains an issue with >20% of patients exhibiting intrinsic resistance and up to 70% of patients developing acquired resistance within 3 years.[1] Many resistance mechanisms converge on the upregulation of CDK6.[2-5] To address this we sought to generate CDK4/6 bifunctional degraders.

METHODS

- PRODEGY platform was utilized to develop a series of cereblon (CRBN) mediated CDK4/6 bifunctional degraders including development candidate BTX-9341.
- Knockout cell lines were generated by nucleofection of Cas9-gRNA complexes.
- Target degradation was analyzed by immunoblots of protein lysates from cells treated with BTX-9341 for 6 hours or as indicated.
- Phosphorylated RB was analyzed by in cell western after 24 hours of treatment or by immunoblot where indicated.
- Cell cycle analysis was performed after 24 hours of treatment by flow cytometry following propidium iodide staining.
- E2F target gene expressed was analyzed by qPCR.
- Cell proliferation was measured by CellTiter-Glo 2.0 assay (Promega) after a 10-day colony formation assay.
- Vehicle, CDK4/6 inhibitor(s), and BTX-9341 were dosed orally in BALB/c nude mice xenograft subcutaneous models.

RESULTS

- BTX-9341 is a potent, CRBN and proteasome dependent degrader of CDK4 and CDK6 in multiple breast cancer cell lines. CDK4/6 degradation is rapid and sustained after compound washout.
- BTX-9341 exhibits a favorable safety profile in THLE2 cells and PBMCs with high μM IC_{50} values.
- Kinome profiling indicates BTX-9341 is more selective than the CDK4/6i palbociclib at 100 nM, and proteomics indicates minimal off-target degradation.
- BTX-9341 functionally inhibits cell proliferation more potently than CDK46i in multiple breast cancer cell lines with IC_{50}s in the low nanomolar range. This enhanced efficacy is CRBN dependent.
- BTX-9341 inhibits RB phosphorylation in breast cancer cells with pRB IC_{50}s below 50 nM.
- BTX-9341 induces cell cycle arrest at low nanomolar concentrations in breast cancer cells.
- BTX-9341 induces potent downregulation of E2F target genes that is sustained over 72 hours.
- BTX-9341 retains potency in a CDK4/6i resistant cell line with CDK6 upregulation.
- BTX-9341 exhibits good tumor exposure when dosed orally, and induces a dose-dependent reduction in CDK4, CDK6, and pRB levels in MCF7 xenograft tumors.
- BTX-9341 exhibits dose dependent tumor growth inhibition and tumor regression at higher doses in an MCF7 xenograft model that was well correlated with CDK4, CDK6 and pRB downregulation.
- BTX-9341 inhibited tumor growth in several HR+/Her2- xenograft models

BTX-9341 exhibits rapid, potent and sustained CDK4 and CDK6 degradation that is CRBN and proteasome dependent

Degradation of CDK4 and CDK6 at 6 hours of treatment in MDA-MB-231 cells

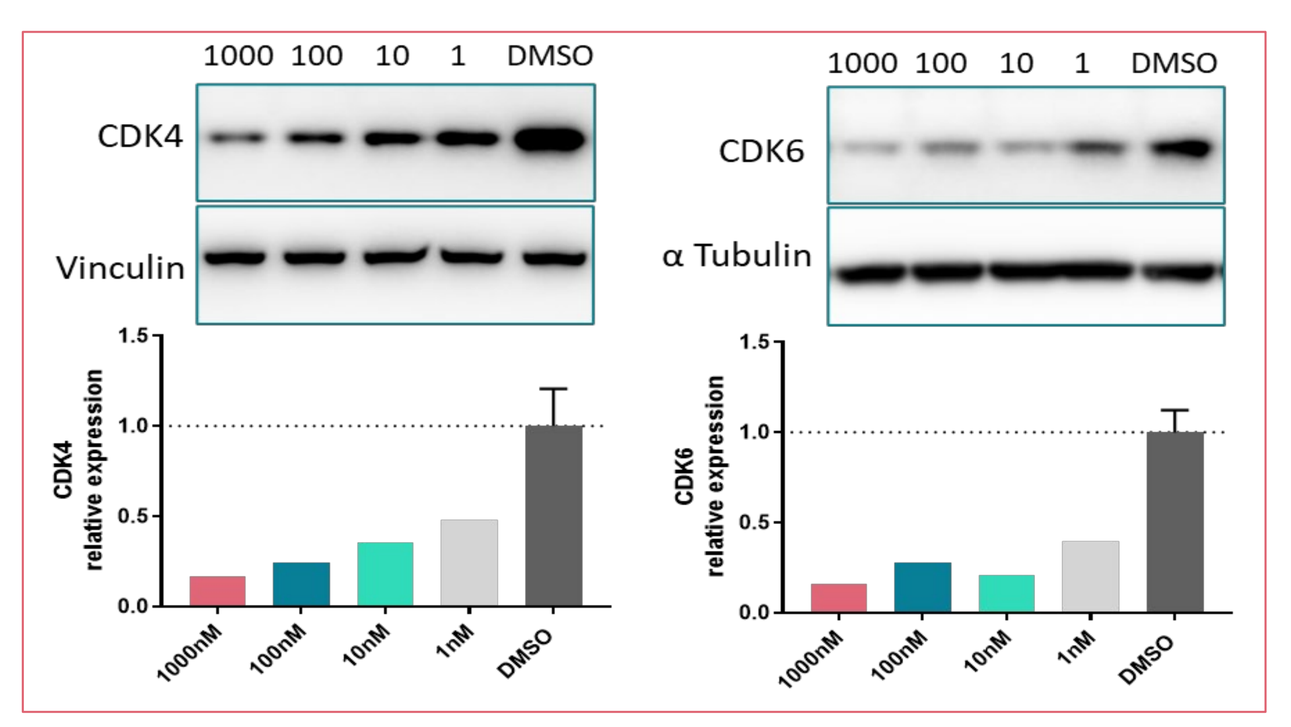
1000 100 10 1 DMSO
CDK4
Vinculin
CDK6
α Tubulin
CDK4 relative expression
CDK6 relative expression
1000nM 100nM 10nM 1nM DMSO

Degradation of CDK4 and CDK6 at 6 hours of treatment in MCF7 and T47D cells

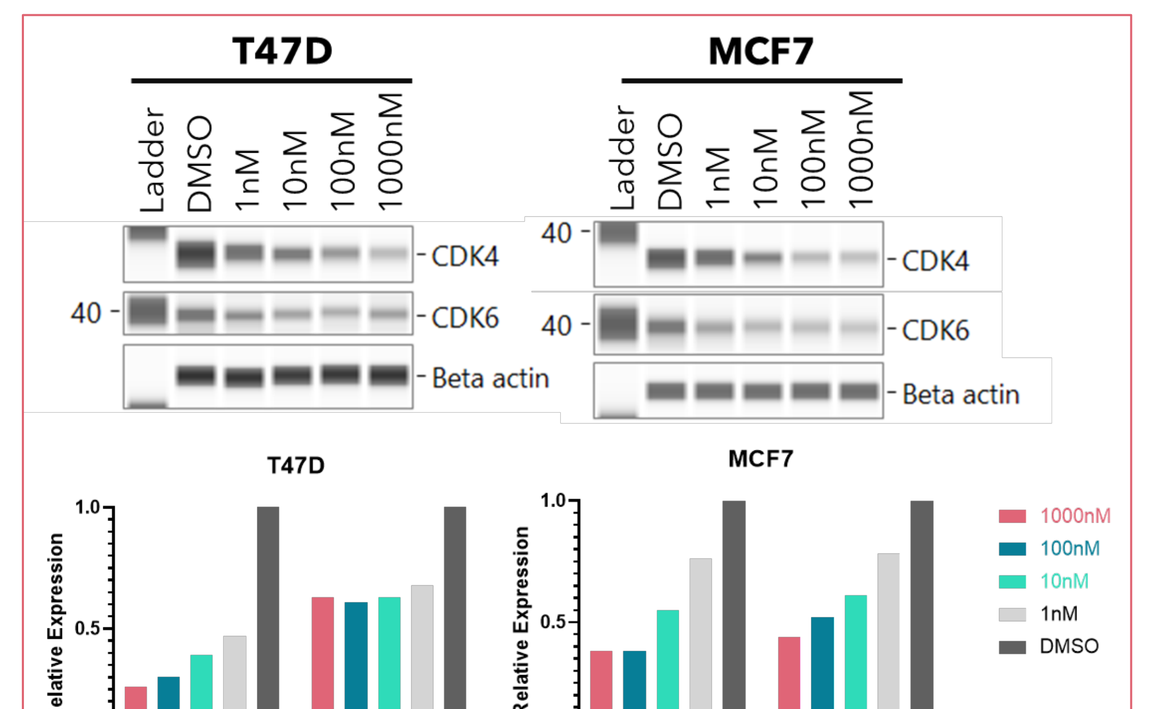
T47D
MCF7
Ladder DMSO 1nM 10nM 100nM 1000nM
CDK4
CDK6
Beta actin
Relative Expression
1000nM 100nM 10nM 1nM DMSO

Degradation of CDK4 and CDK6 at 6 hours of treatment in MDA-MB-231 control and CRBN knockout cells

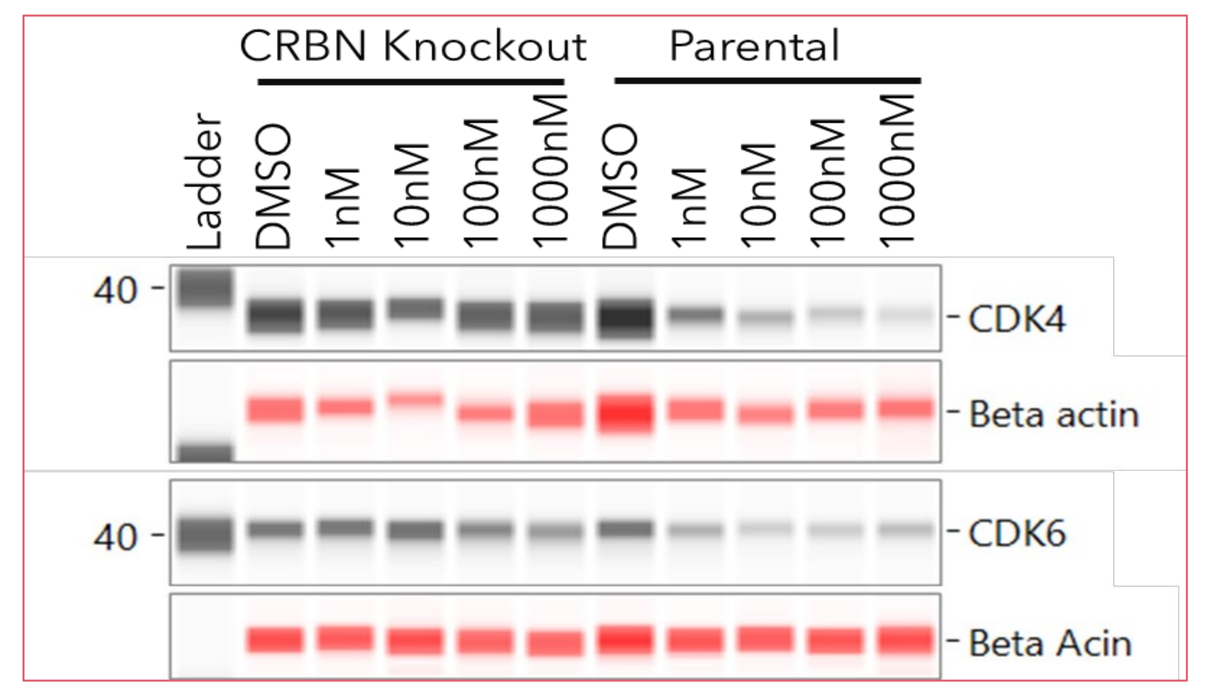
CRBN Knockout
Parental
Ladder DMSO 1nM 10nM 100nM 1000nM DMSO 1nM 10nM 100nM 1000nM
CDK4
Beta actin
CDK6
Beta Acin

Degradation of CDK4 and CDK6 at 6 hours of treatment in MDA-MB-231 cells with MG132

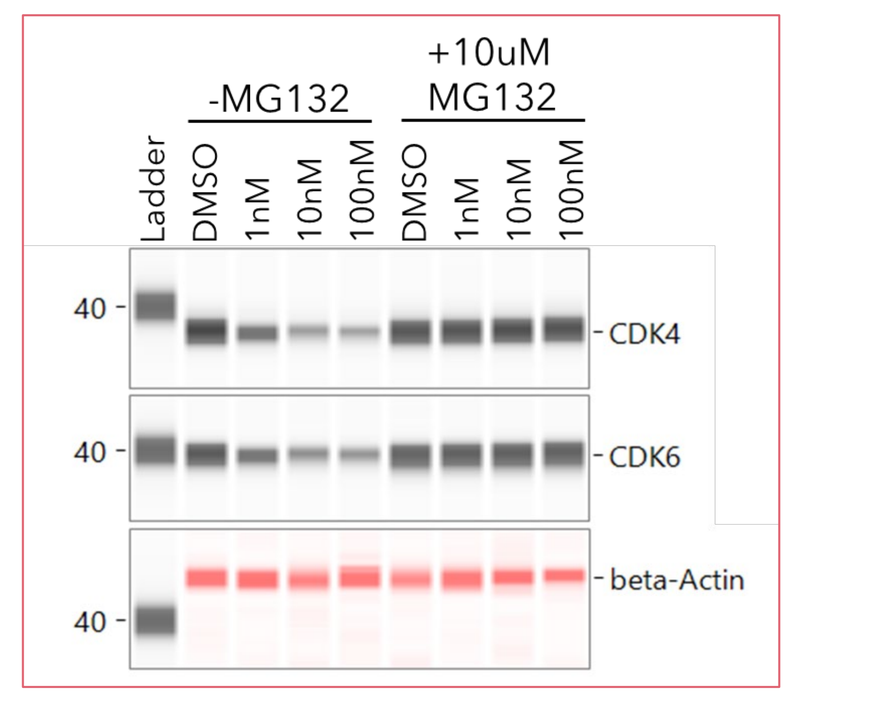
-MG132
+10uM MG132
CDK4
CDK6
beta-Actin

Degradation of CDK4 and CDK6 at 2,4, and 6 hours of treatment in MDA-MB-231 cells

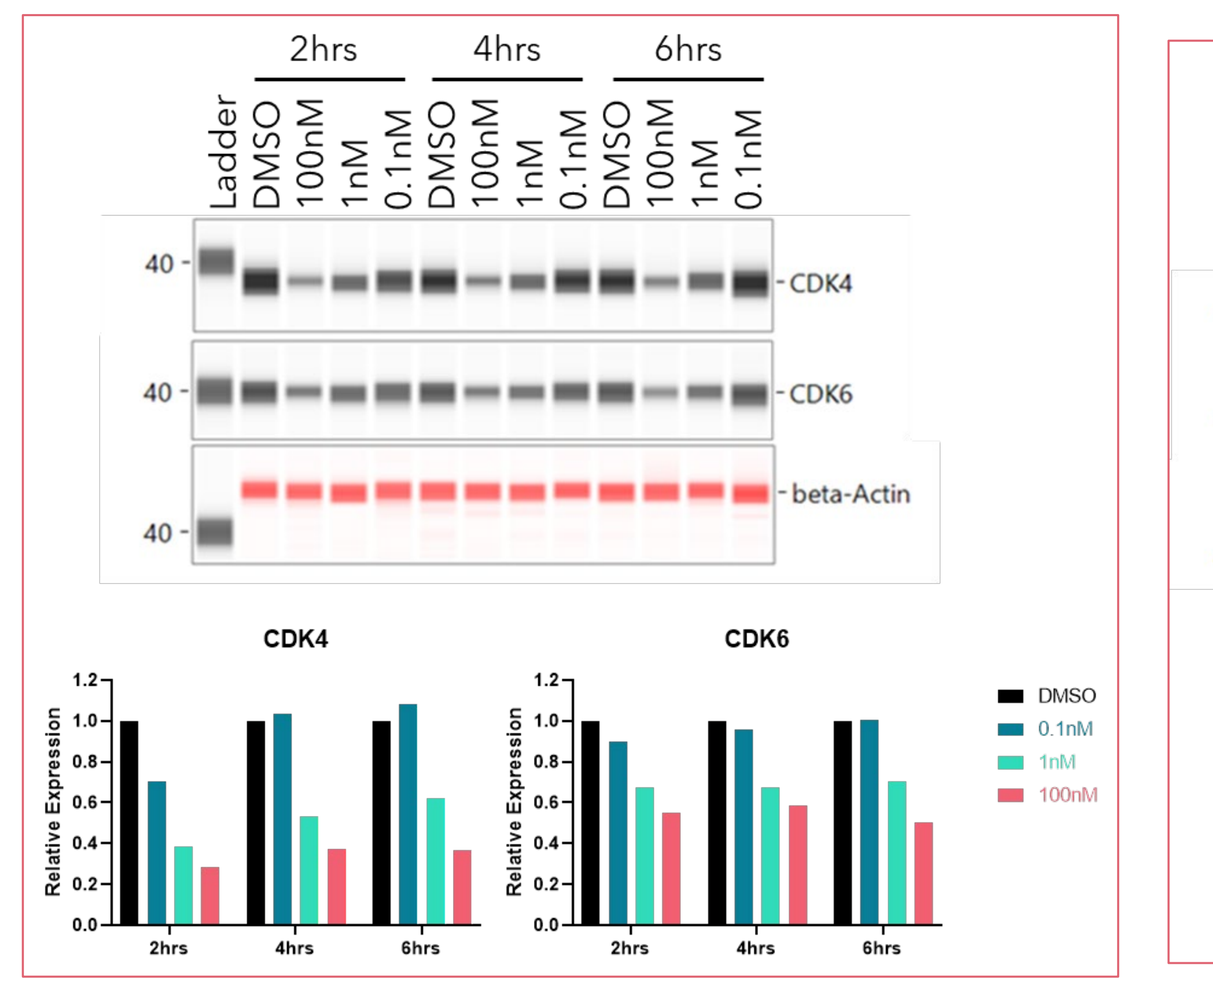
2hrs
4hrs
6hrs
CDK4
CDK6
beta-Actin
Relative Expression
DMSO
0.1nM
1nM
100nM

CDK4 and CDK6 protein recovery 8, 24 and 48 hours after washout of 16 hours treatment in MDA-MB-231 cells

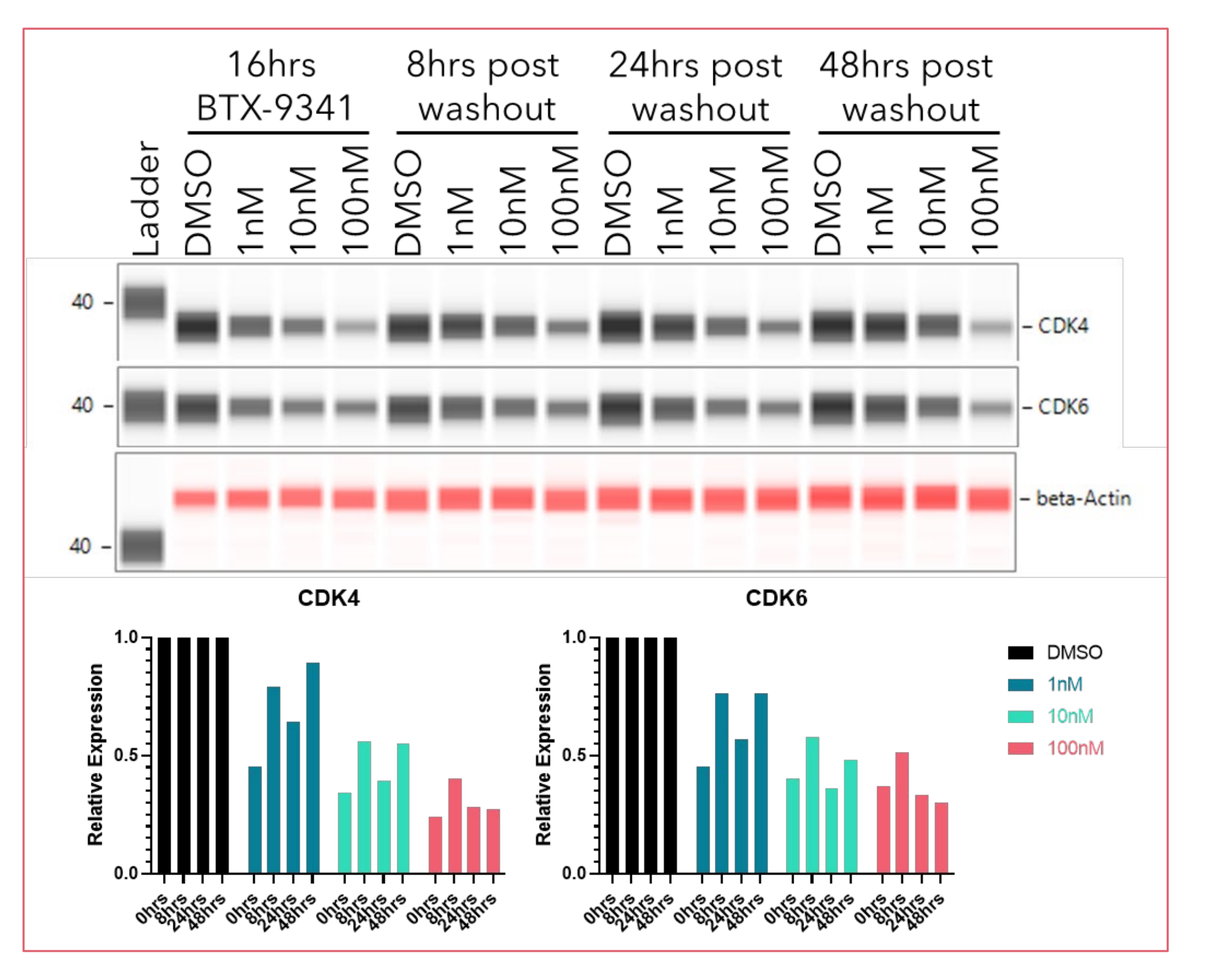
16hrs BTX-9341
8hrs post washout
24hrs post washout
48hrs post washout
CDK4
CDK6
beta-Actin
Relative Expression
DMSO
1nM
10nM
100nM

BTX-9341 exhibits a favorable safety profile in PBMCs and THLE2 cells

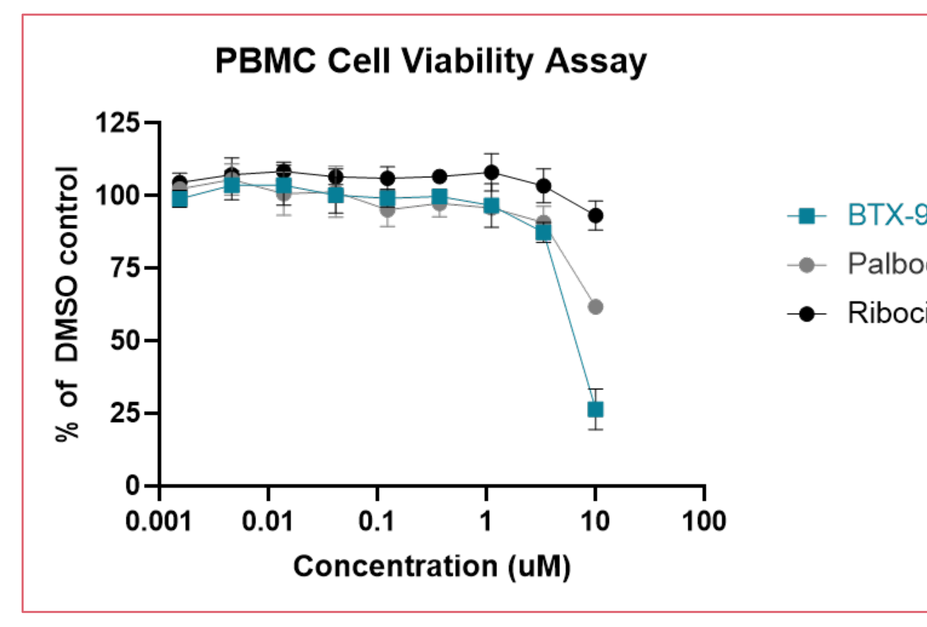
PBMC Cell Viability Assay
% of DMSO control
Concentration (uM)
BTX-9341
Palbociclib
Ribociclib

PBMCs treated with compound for 24hrs

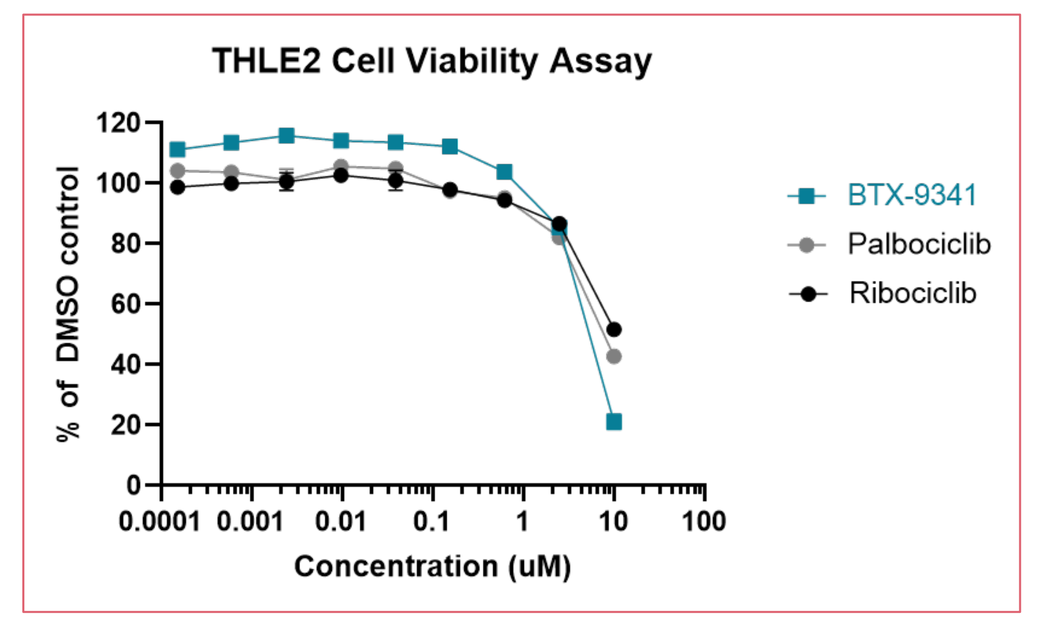
THLE2 Cell Viability Assay
% of DMSO control
Concentration (uM)
BTX-9341
Palbociclib
Ribociclib

THLE2 cells treated with compound for 72hrs

BTX-9341 exhibits selective binding and degradation

Compound	Kinome S(35) score @100nM
BTX-9341	0.01
Palbociclib	0.027

Compound	CDK6 Kd
BTX-9341	31 nM
Palbociclib	5.1 nM

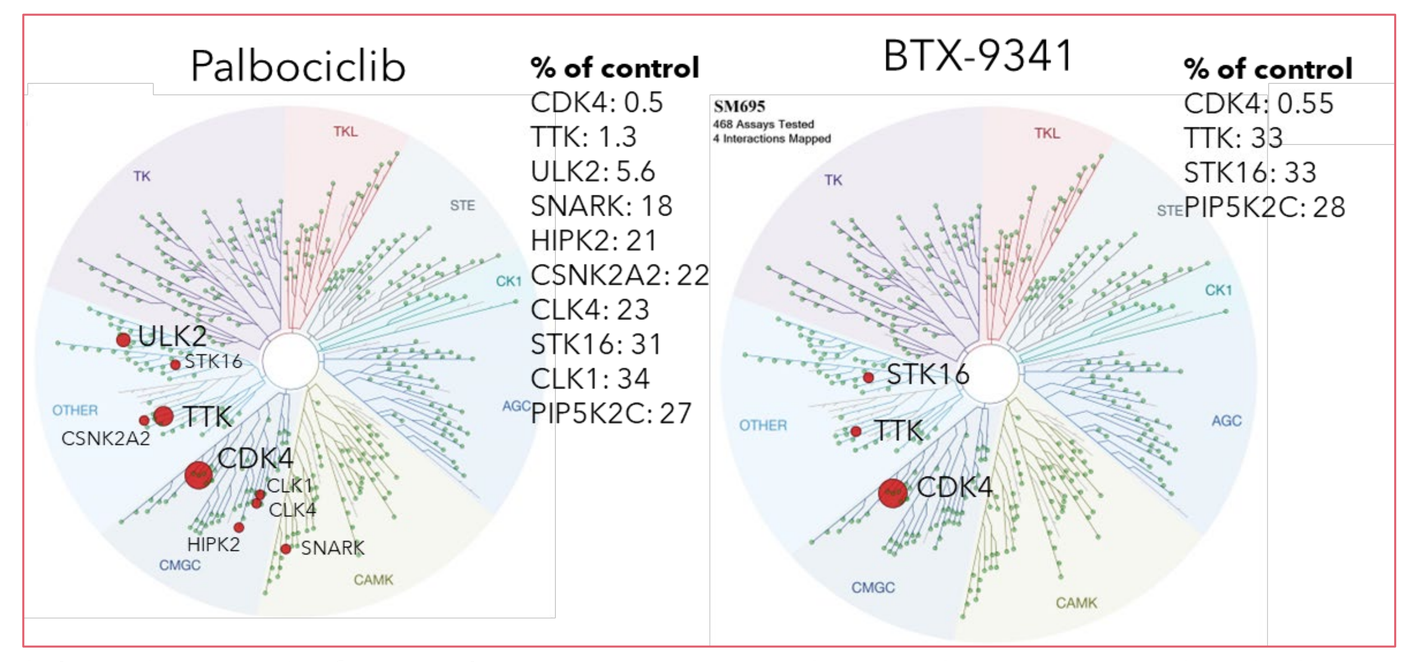
Palbociclib
% of control
CDK4: 0.5
TTK: 1.3
ULK2: 5.6
SNARK: 18
HIPK2: 21
CSNK2A2: 22
CLK4: 23
STK16: 31
CLK1: 34
PIP5K2C: 27
BTX-9341
% of control
CDK4: 0.55
TTK: 33
STK16: 33
PIP5K2C: 28

Kinome S(35) score: the fraction of kinases with less than 35% of control at 100nM among the 468 kinases tested with the KINOMEscan™ platform from Eurofins.

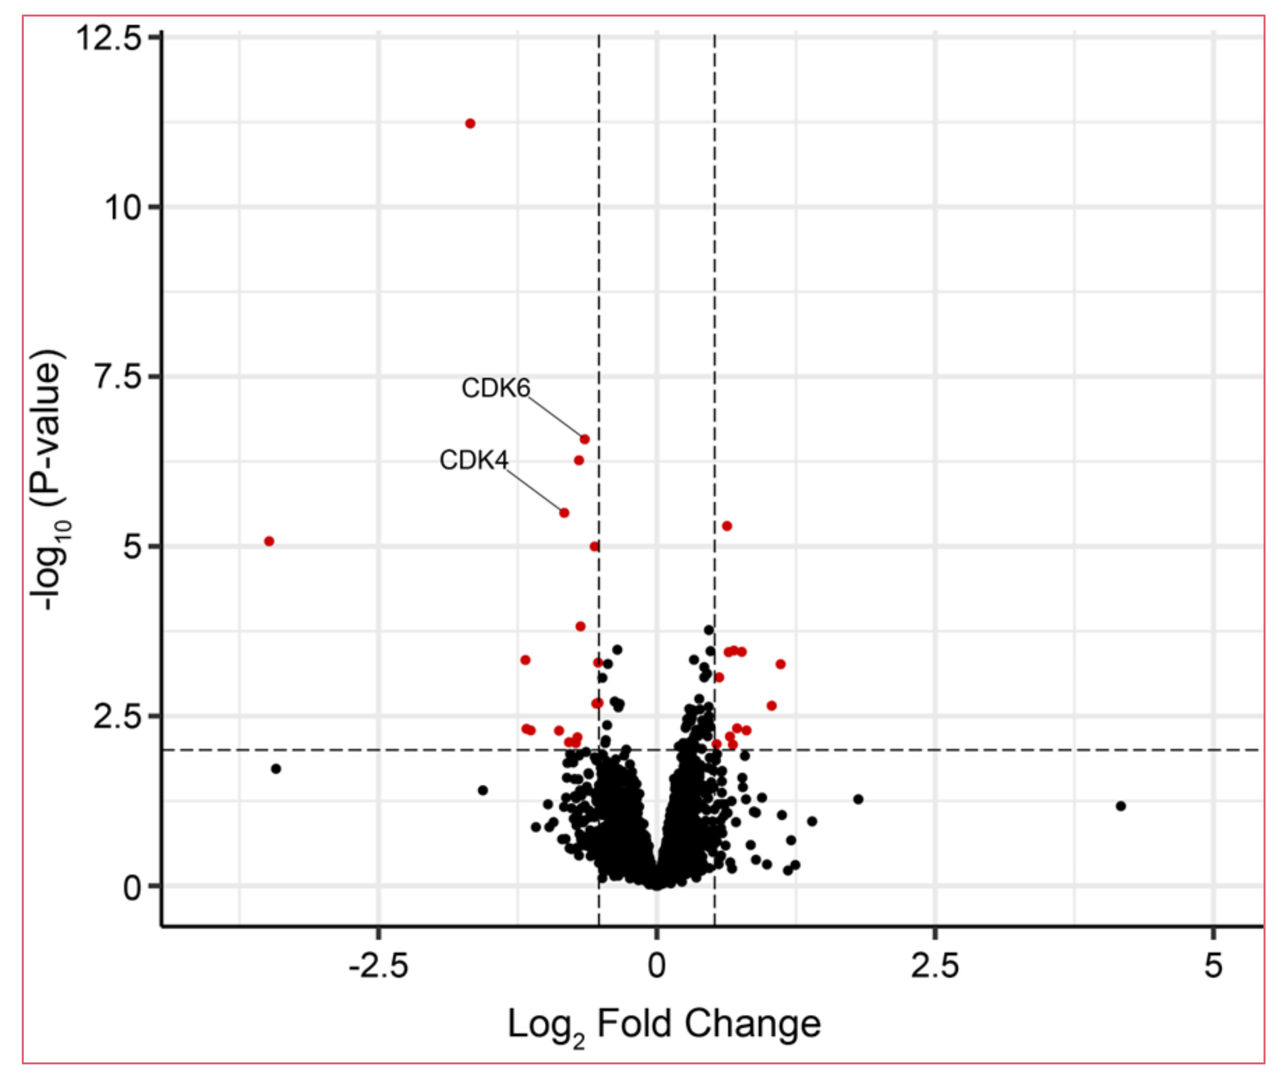
-log_{10} (P-value)
Log_2 Fold Change
CDK6
CDK4

TMT mass spectrometry performed on samples generated in triplicate from MDA-MB-231 cells treated with 100 nM of BTX-9341 or DMSO for 6 hours.

Volcano plot indicating the fold change comparing BTX-9341 treated and DMSO treated cells. CDK4 and CDK6 are labeled as some of the few proteins which were significantly altered after BTX-9341 treatment.

BTX-9341 potently inhibits downstream signaling and cell proliferation *in vitro* in HR+/HER2- BC cells, CDK4/6i resistant cells and TNBC

BTX-9341 inhibits pRB in BC cells

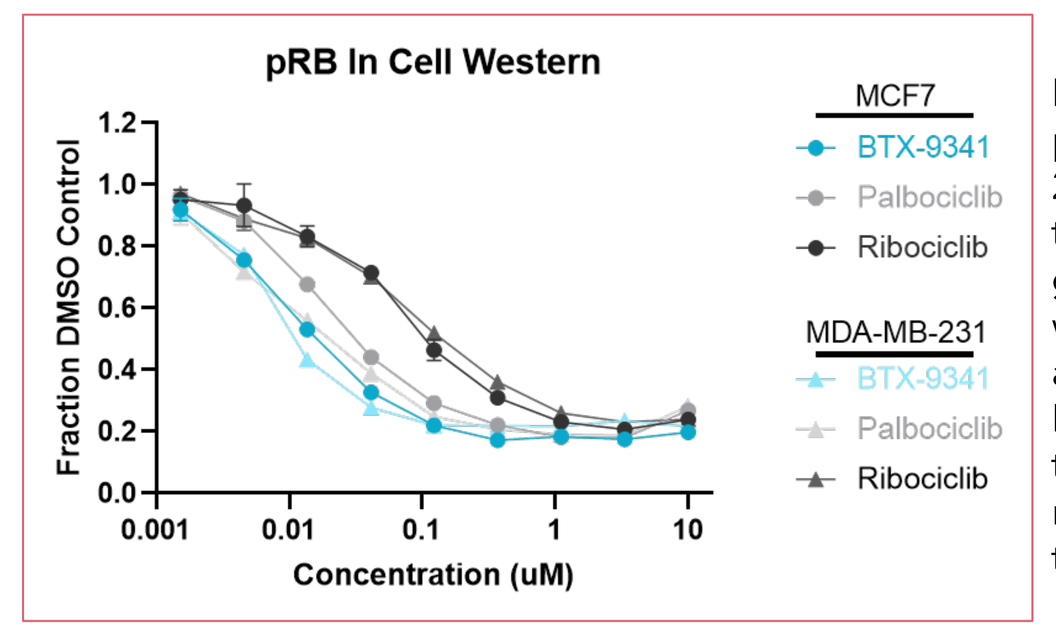
pRB In Cell Western
Fraction DMSO Control
Concentration (uM)
MCF7
BTX-9341
Palbociclib
Ribociclib
MDA-MB-231
BTX-9341
Palbociclib
Ribociclib

BTX-9341 inhibits RB phosphorylation after 24 hours of treatment. Data generated by in cell western for total RB and phosphorylated RB (pSer807/811) taken as a ratio and normalized to DMSO treated control.

BTX-9341 inhibits cell proliferation in a CRBN dependent manner

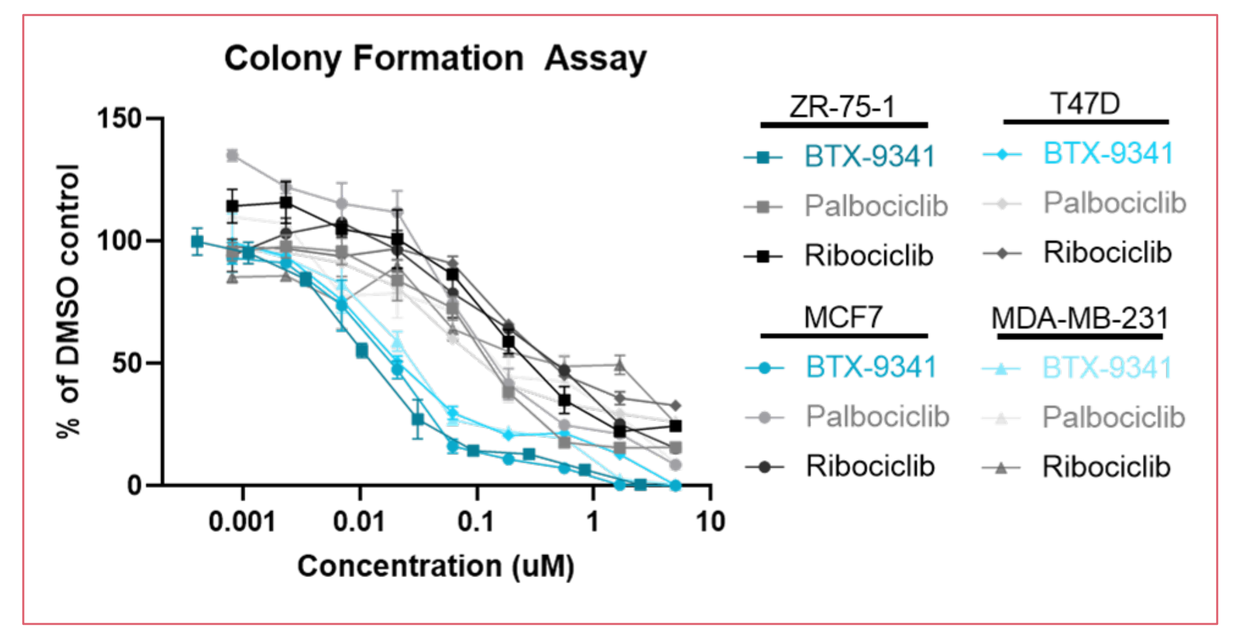
Colony Formation Assay
% of DMSO control
Concentration (uM)
ZR-75-1
T47D
MCF7
MDA-MB-231
BTX-9341
Palbociclib
Ribociclib

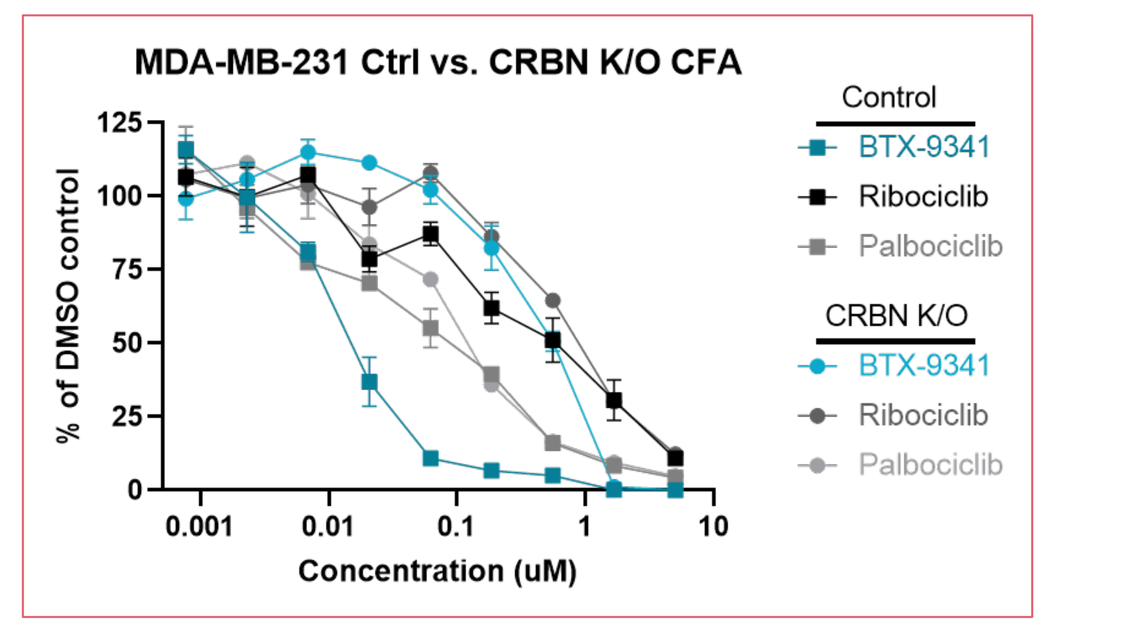
MDA-MB-231 Ctrl vs. CRBN K/O CFA
% of DMSO control
Concentration (uM)
Control
CRBN K/O
BTX-9341
Ribociclib
Palbociclib

BTX-9341 inhibits cell proliferation and RB phosphorylation in a palbociclib resistant cell line

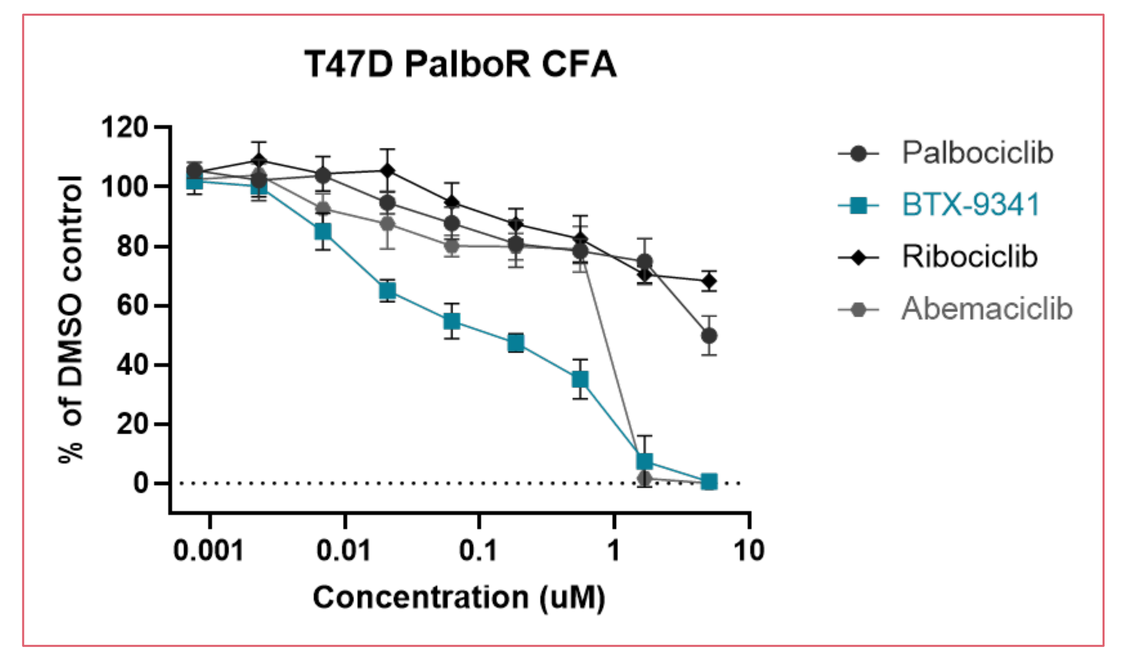
T47D PalboR CFA
% of DMSO control
Concentration (uM)
Palbociclib
BTX-9341
Ribociclib
Abemaciclib

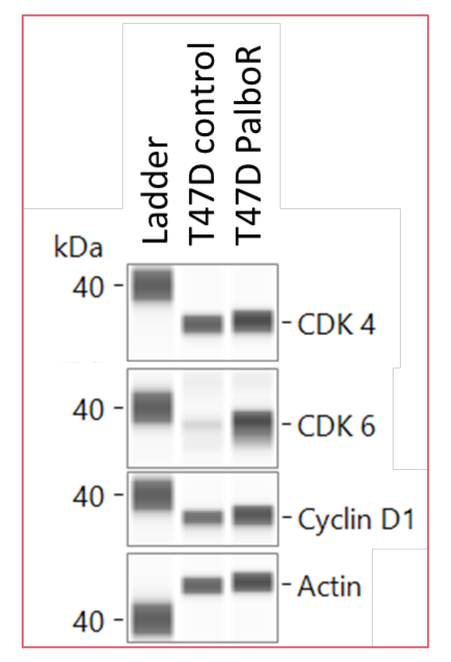
kDa
Ladder
T47D control
T47D PalboR
CDK 4
CDK 6
Cyclin D1
Actin

BTX-9341 induces cell cycle arrest

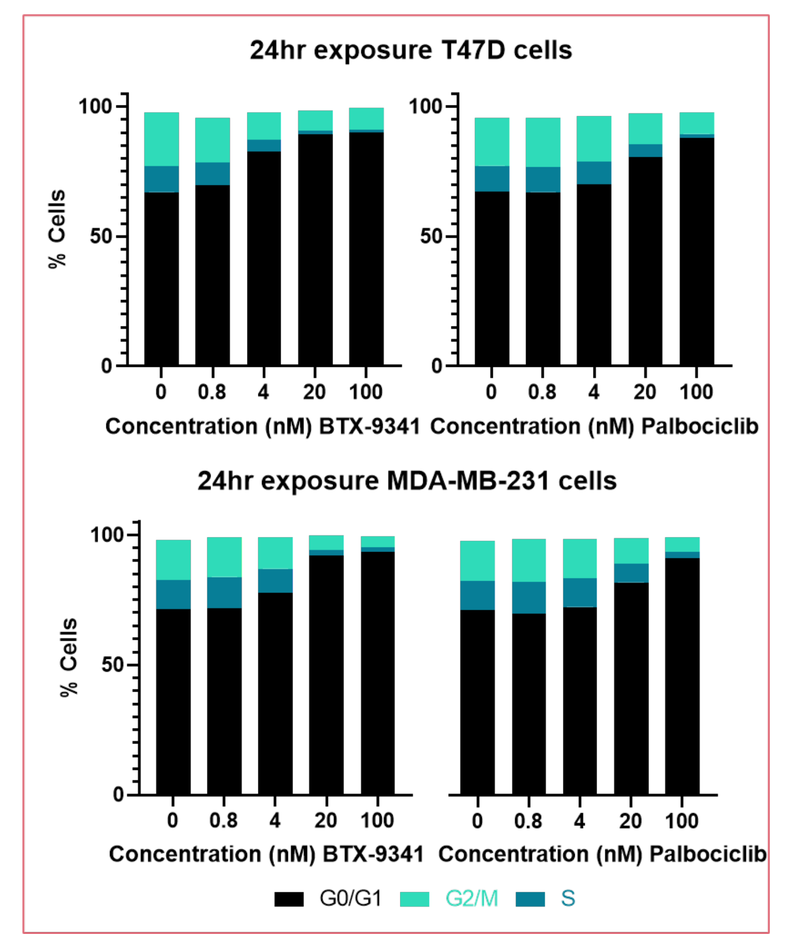
24hr exposure T47D cells
24hr exposure MDA-MB-231 cells
% Cells
Concentration (nM) BTX-9341
Concentration (nM) Palbociclib
G0/G1
G2/M
S

BTX-9341 downregulates E2F target genes

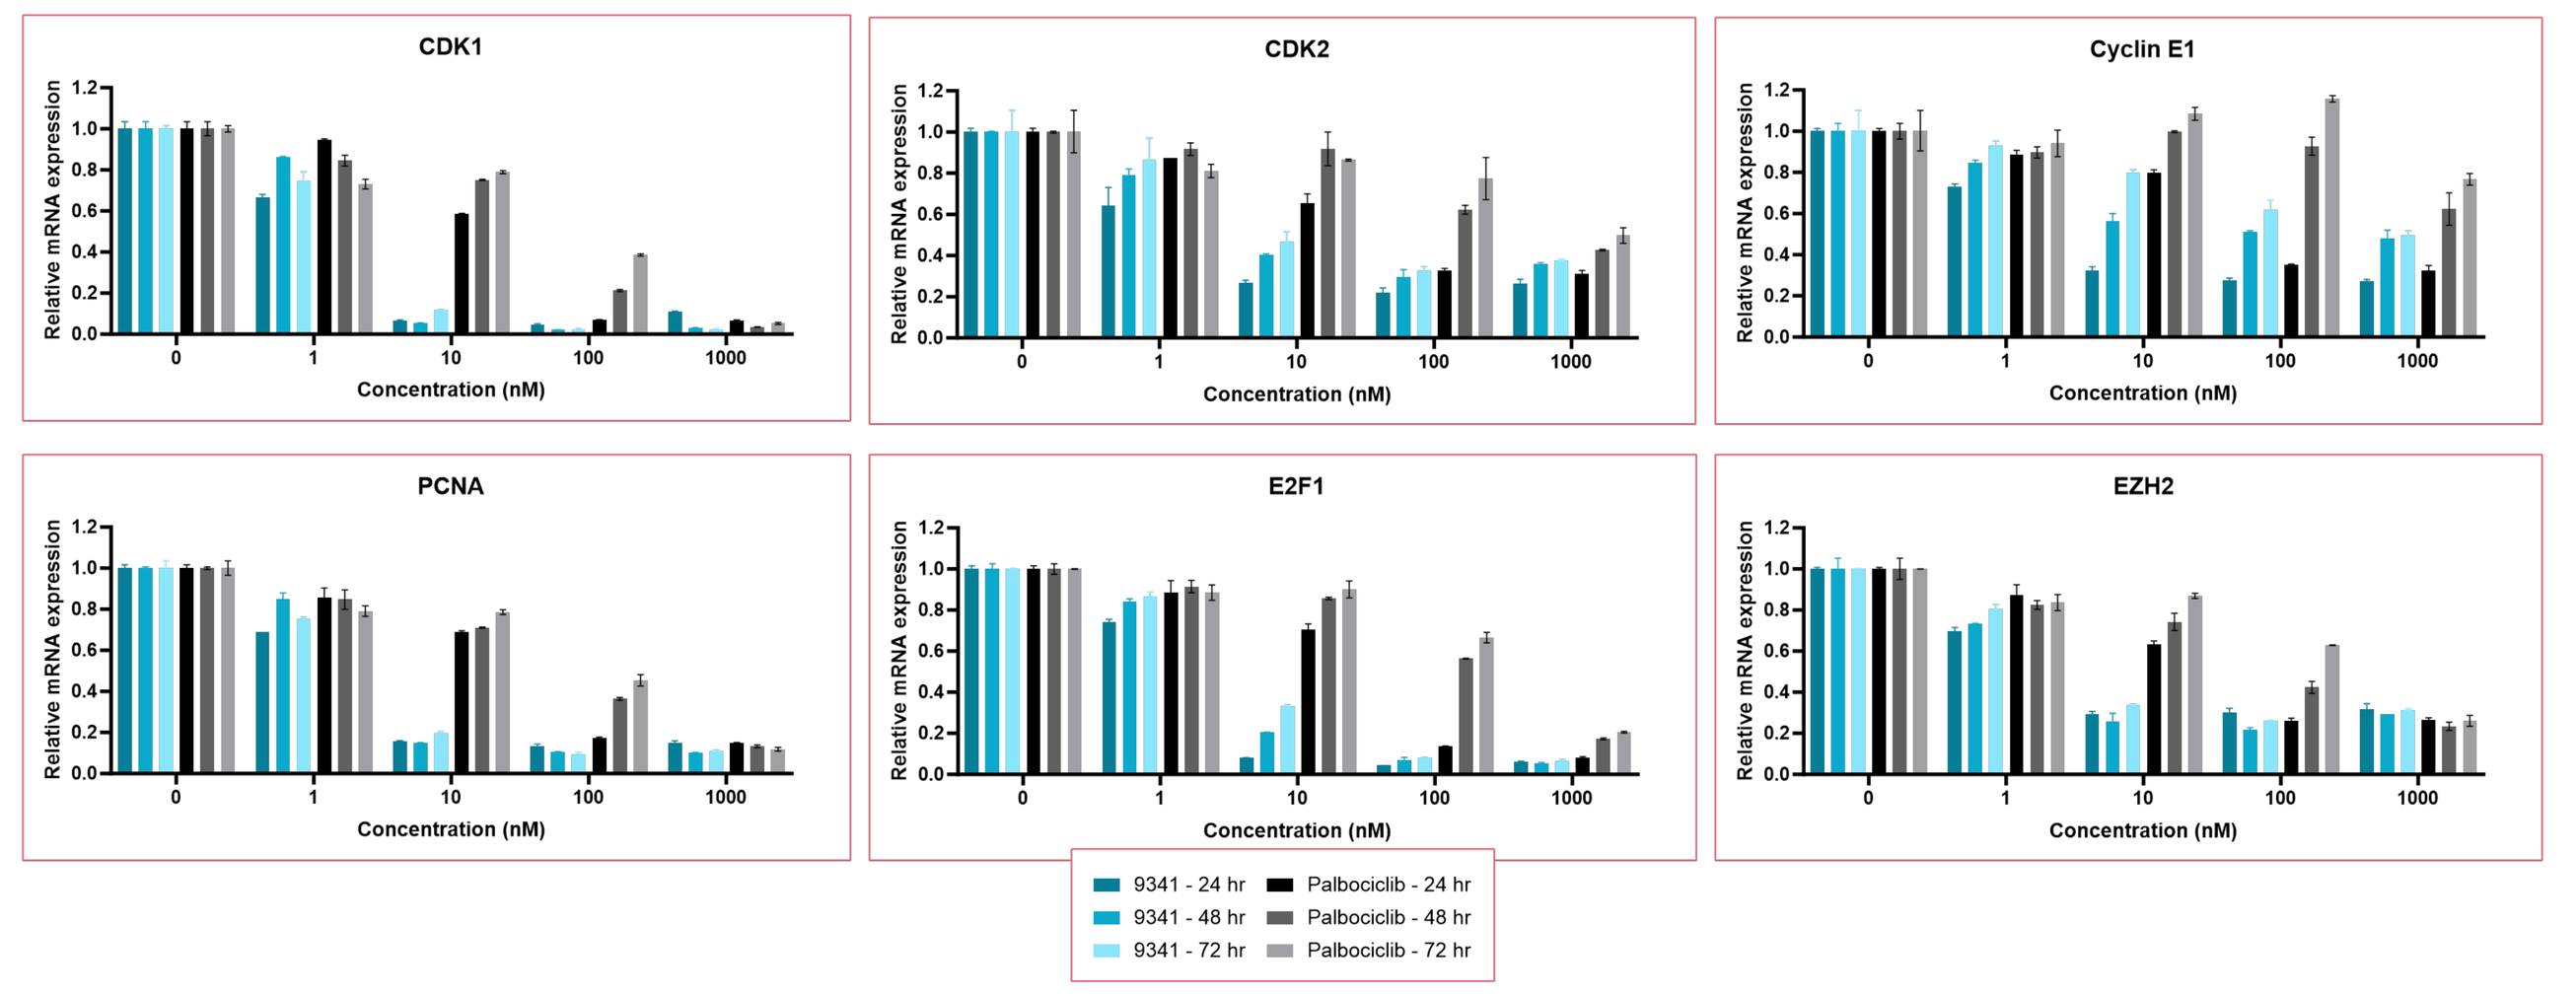
CDK1
CDK2
Cyclin E1
PCNA
E2F1
EZH2
Relative mRNA expression
Concentration (nM)
9341 - 24 hr
9341 - 48 hr
9341 - 72 hr
Palbociclib - 24 hr
Palbociclib - 48 hr
Palbociclib - 72 hr

BTX-9341 inhibits E2F target gene expression in T47D cells. Suppression of gene expression is sustained through 72 hours, while recovery of gene expression is seen starting at 48 hours after palbociclib treatment

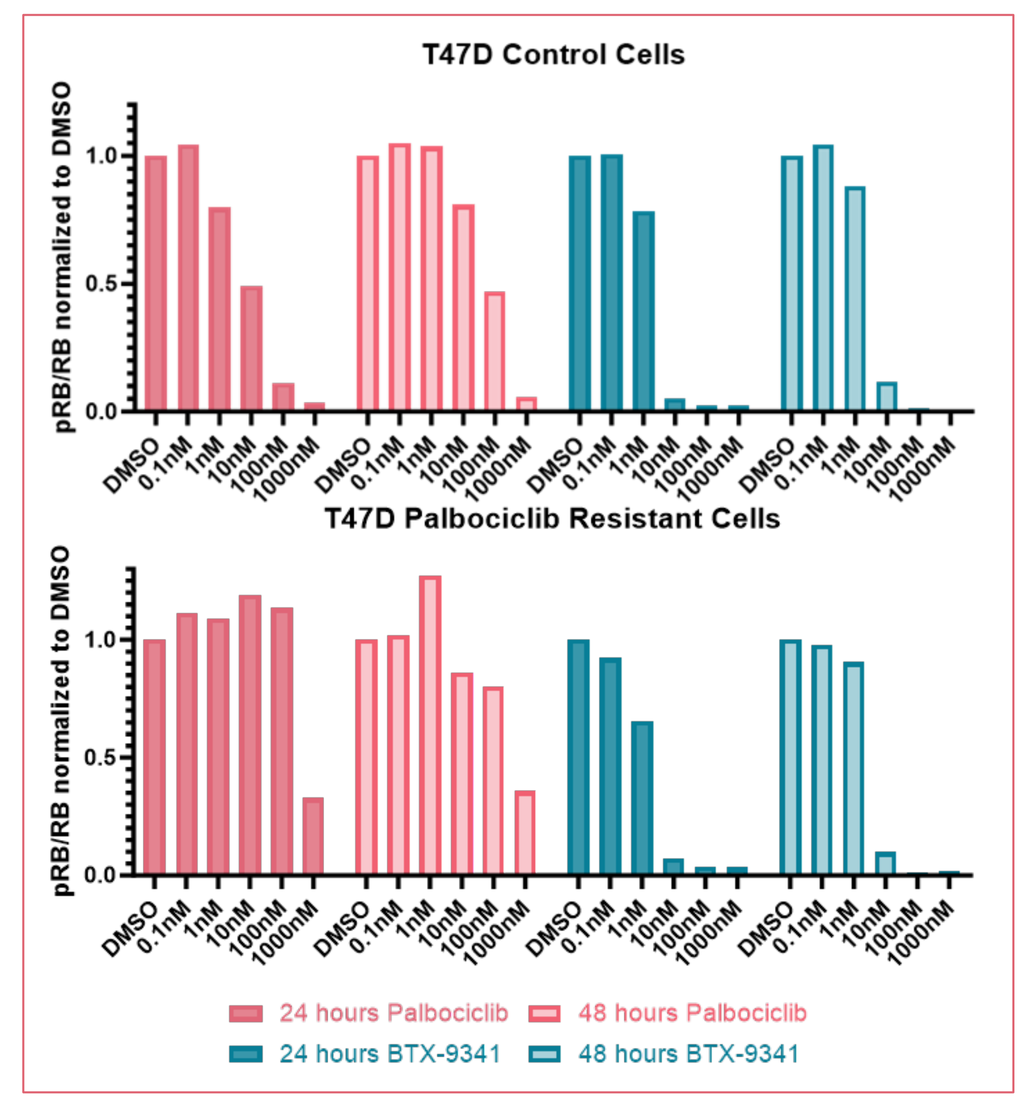
T47D Control Cells
T47D Palbociclib Resistant Cells
pRB/RB normalized to DMSO
24 hours Palbociclib
48 hours Palbociclib
24 hours BTX-9341
48 hours BTX-9341

By immunoblot BTX-9341 inhibits RB phosphorylation in both T47D control and palbociclib resistant cell lines with the same potency. Palbociclib shows minimal inhibition of RB phosphorylation,. BTX-9341 inhibition of pRB is sustained through 48 hours, while pRB begins to recover after 48 hours with palbociclib treatment.

BTX-9341 induces tumor regression in MCF7 xenograft model

BTX-9341 degrades CDK4, and CDK6, AND inhibits pRB in MCF7subcutaneous tumors

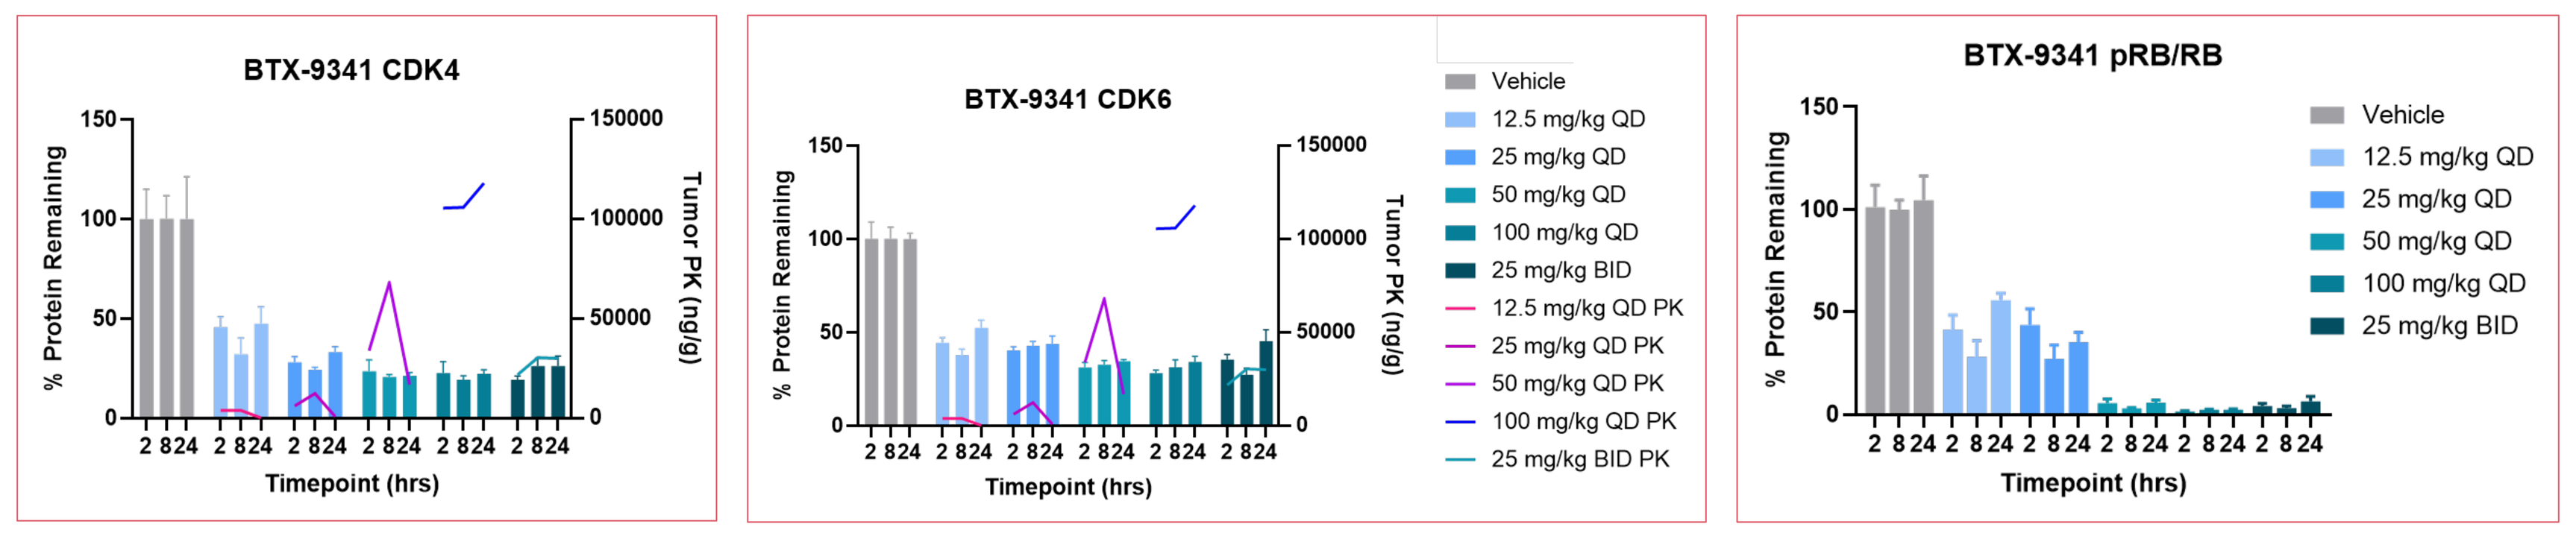
BTX-9341 CDK4
BTX-9341 CDK6
BTX-9341 pRB/RB
% Protein Remaining
Tumor PK (ng/g)
Timepoint (hrs)
Vehicle
12.5 mg/kg QD
25 mg/kg QD
50 mg/kg QD
100 mg/kg QD
25 mg/kg BID
12.5 mg/kg QD PK
25 mg/kg QD PK
50 mg/kg QD PK
100 mg/kg QD PK
25 mg/kg BID PK

BTX-9341 exhibits dose-dependent target degradation of CDK4 and CDK6 which correlates with a dose-dependent decrease in phosphorylated RB relative to total RB that is rapid and sustained.

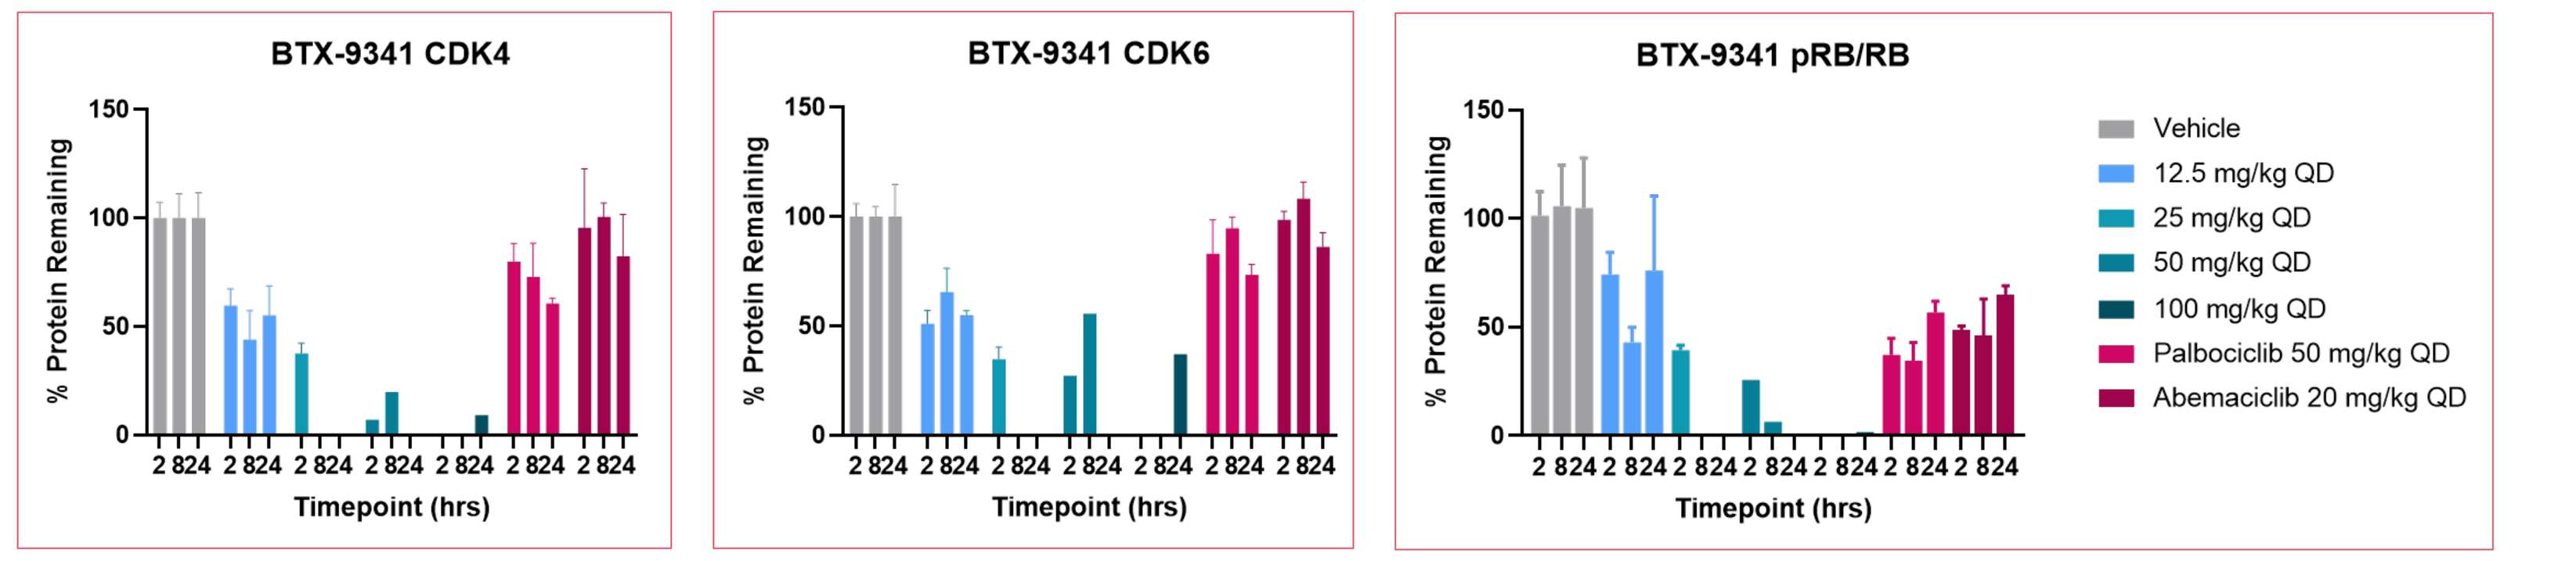
BTX-9341 CDK4
BTX-9341 CDK6
BTX-9341 pRB/RB
% Protein Remaining
Timepoint (hrs)
Vehicle
12.5 mg/kg QD
25 mg/kg QD
50 mg/kg QD
100 mg/kg QD
Palbociclib 50 mg/kg QD
Abemaciclib 20 mg/kg QD

BTX-9341 exhibits dose-dependent target degradation of CDK4 and CDK6 which correlates with a dose-dependent decrease in phosphorylated RB relative to total RB in and MCF7 xenograft efficacy model. Decreases in pRB more significant than CDK4/6i at dose levels higher than 25mpk.

BTX-9341 induces tumor regression in an MCF7 xenograft model with multiple dosing schedules

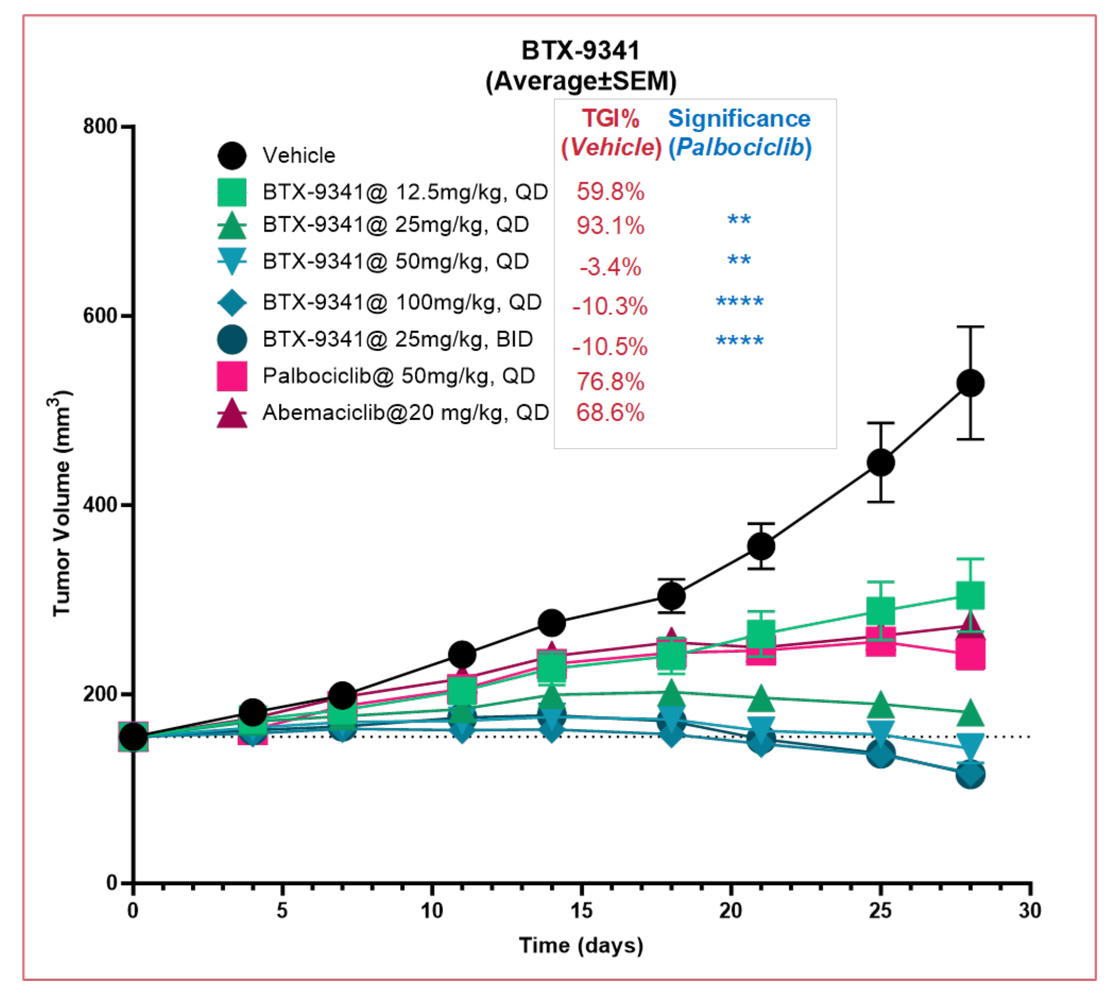
BTX-9341
(Average±SEM)
TGI% (Vehicle)
Significance (Palbociclib)
Vehicle
BTX-9341@ 12.5mg/kg, QD 59.8%
BTX-9341@ 25mg/kg, QD 93.1% **
BTX-9341@ 50mg/kg, QD -3.4% **
BTX-9341@ 100mg/kg, QD -10.3% ****
BTX-9341@ 25mg/kg, BID -10.5% ****
Palbociclib@ 50mg/kg, QD 76.8%
Abemaciclib@20 mg/kg, QD 68.6%
Tumor Volume (mm^3)
Time (days)

BTX-9341 exhibits dose-dependent tumor growth inhibition in an MCF7 xenograft model. BTX-9341 dosed at 25mpk BID, 50 mpk QD and 100 mpk QD all cause tumor regression in this model.

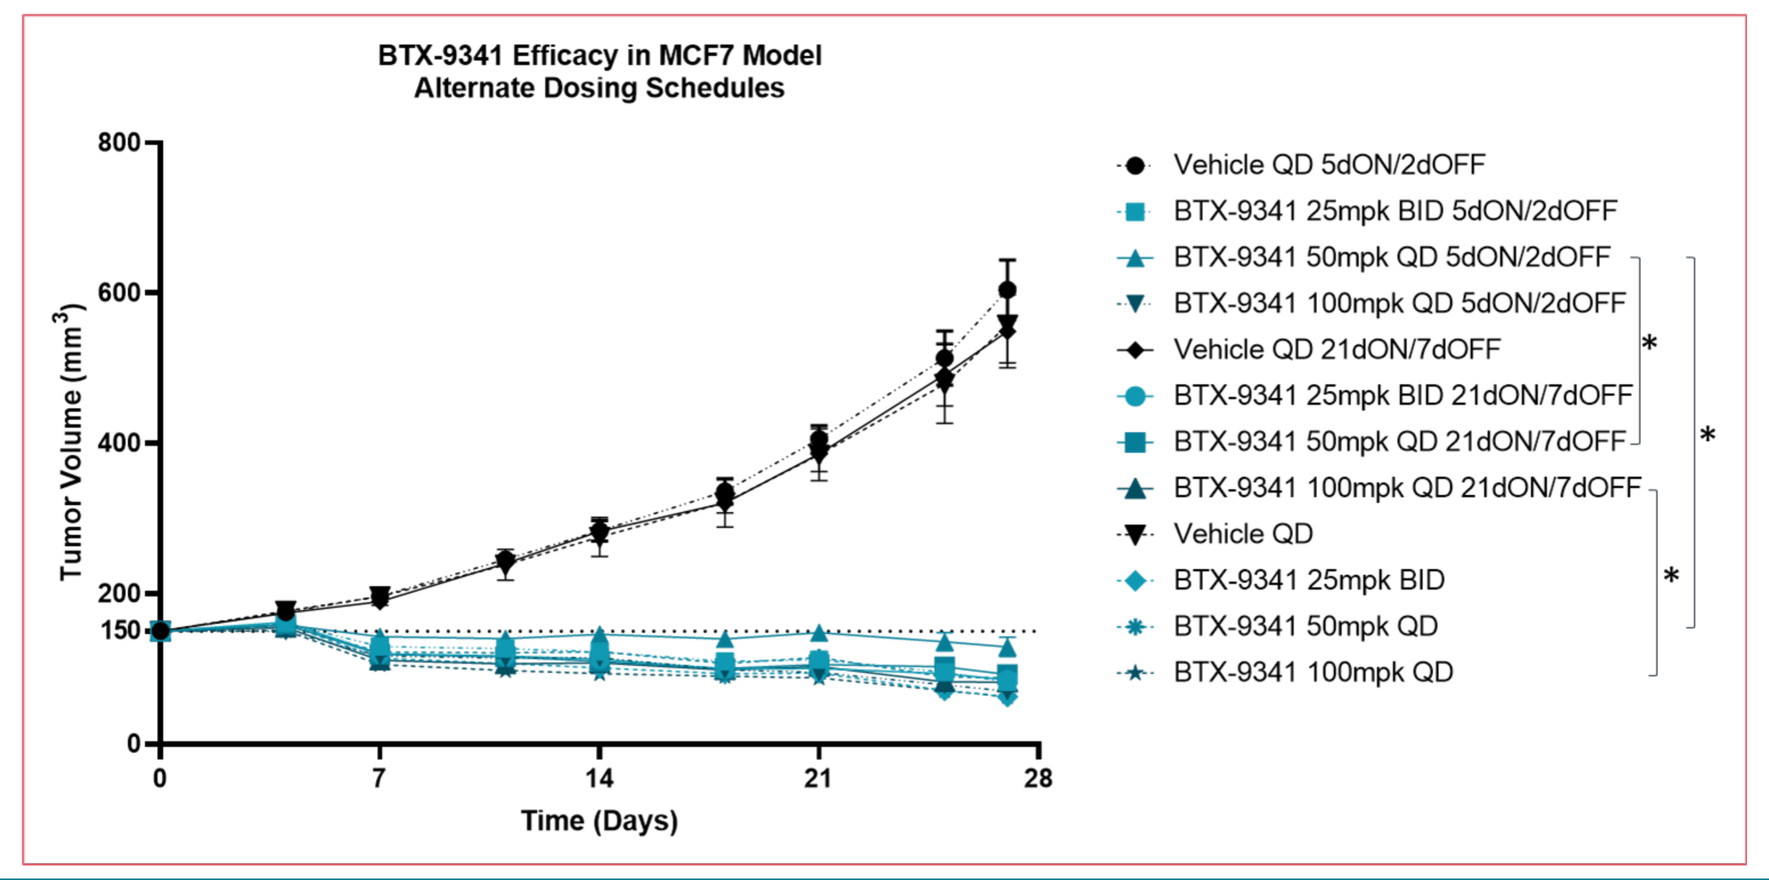
BTX-9341 Efficacy in MCF7 Model
Alternate Dosing Schedules
Tumor Volume (mm^3)
Time (Days)
Vehicle QD 5dON/2dOFF
BTX-9341 25mpk BID 5dON/2dOFF
BTX-9341 50mpk QD 5dON/2dOFF
BTX-9341 100mpk QD 5dON/2dOFF
Vehicle QD 21dON/7dOFF
BTX-9341 25mpk BID 21dON/7dOFF
BTX-9341 50mpk QD 21dON/7dOFF
BTX-9341 100mpk QD 21dON/7dOFF
Vehicle QD
BTX-9341 25mpk BID
BTX-9341 50mpk QD
BTX-9341 100mpk QD

BTX-9341 induces tumor regression in an MCF7 xenograft model at 25mpk BID, 50mpk QD and 100 mpk QD in a 5 day on, 2 day off dosing schedule and a 21 day on 7 day off dosing schedule with minor but significant differences in potency compared to a full 28-day dosing for dose levels except 25 mpk BID.

BTX-9341 inhibits tumor growth and induces tumor regression in multiple HR+/Her2- xenograft models

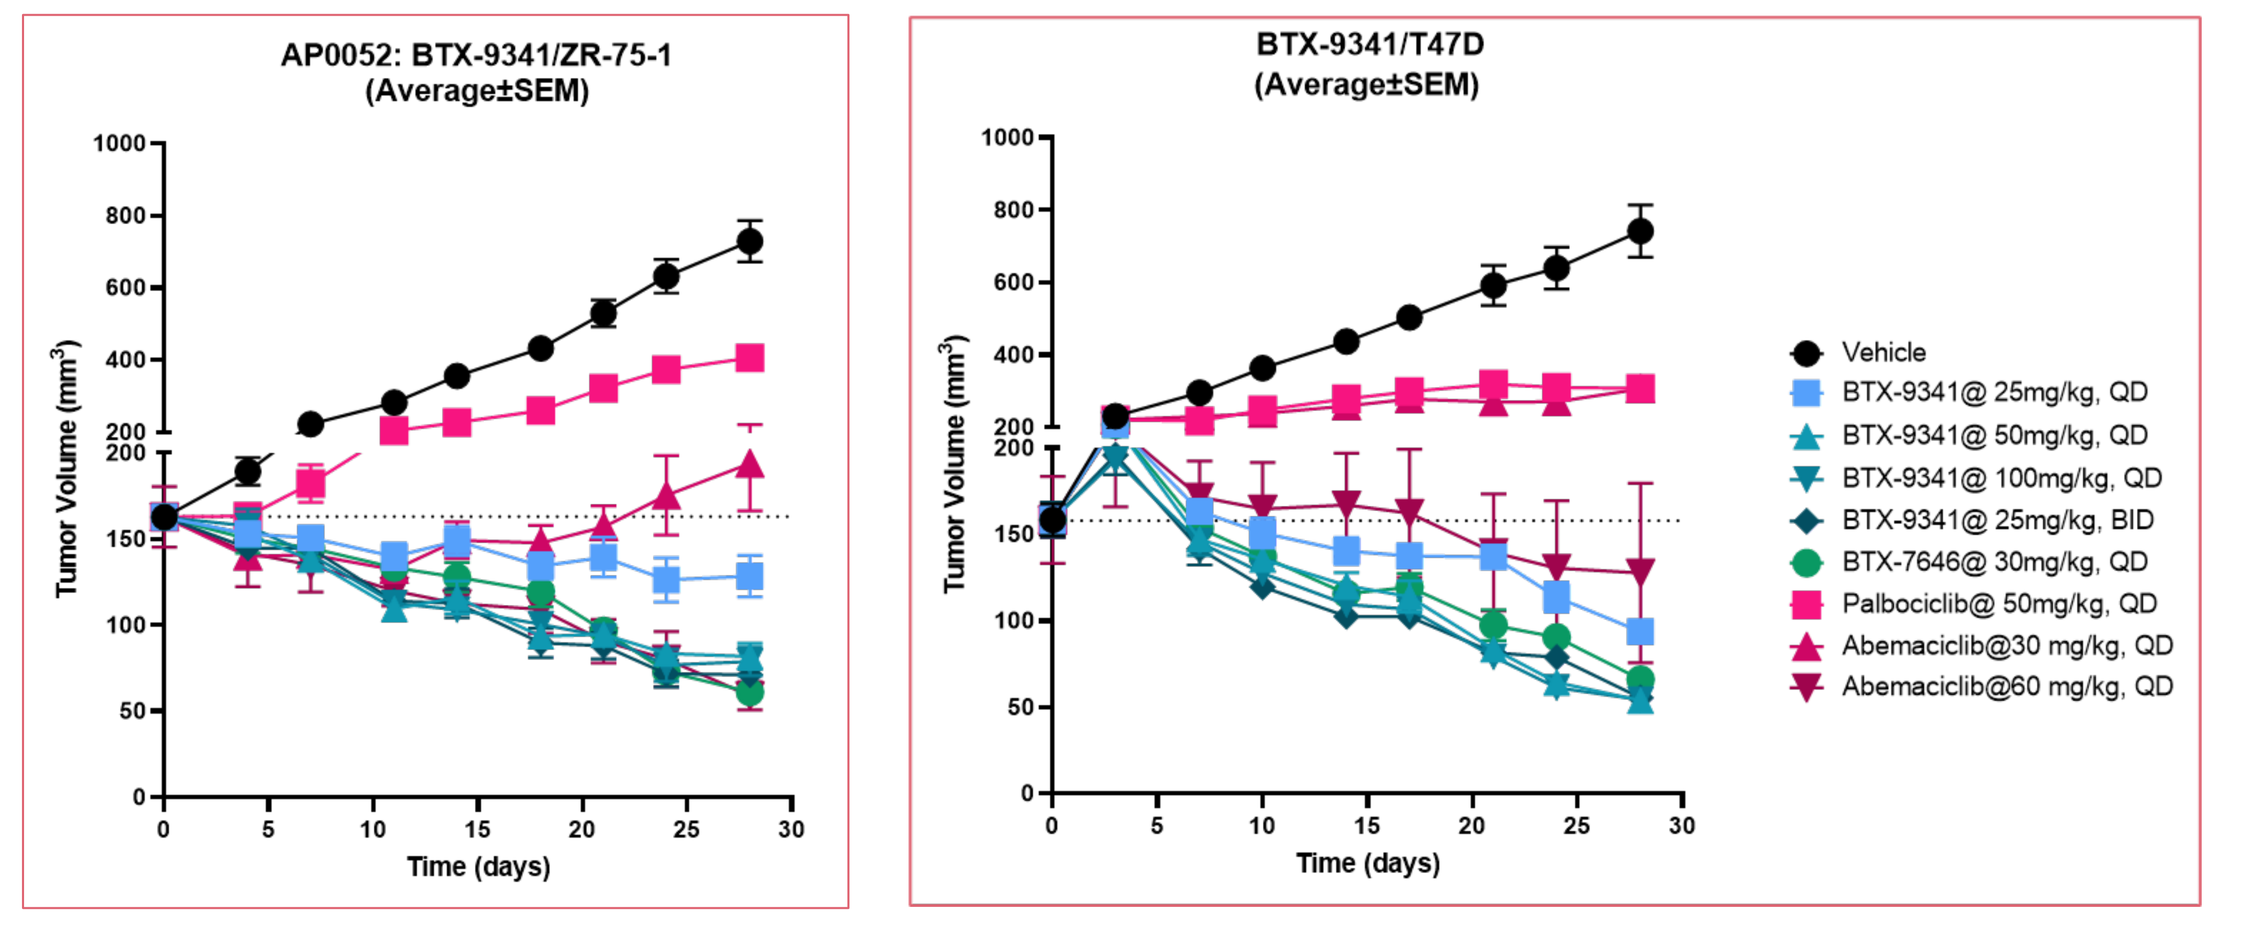
AP0052: BTX-9341/ZR-75-1
(Average±SEM)
BTX-9341/T47D
(Average±SEM)
Tumor Volume (mm^3)
Time (days)
Vehicle
BTX-9341@ 25mg/kg, QD
BTX-9341@ 50mg/kg, QD
BTX-9341@ 100mg/kg, QD
BTX-9341@ 25mg/kg, BID
BTX-7646@ 30mg/kg, QD
Palbociclib@ 50mg/kg, QD
Abemaciclib@30 mg/kg, QD
Abemaciclib@60 mg/kg, QD

BTX-9341 exhibits dose-dependent tumor growth inhibition in two HR+/HER2- xenograft models ZR-75-1 and T47D. Tumor growth inhibition at all doses of BTX-9341 is more potent than palbociclib and a lower dose of Abemaciclib, and in the T47D model, all doses are more potent than this higher dose of abemaciclib as well. Tumor regression is seen at all doses of BTX-9341 tested.

CONCLUSIONS

These preclinical data show that BTX-9341 promotes CRBN and proteasome dependent degradation of CDK4 and CDK6 in multiple breast cancer cell lines. This degradation is specific, with limited off-target binding and degradation. This degradation leads to a more potent phenotype in *in vitro* compared to CDK4/6i with a deeper and more sustained inhibition of cell cycle progression, phospho-RB, and E2F target gene expression. BTX-9341 exhibited more potent tumor growth inhibition in multiple HR+/HER2- xenograft models compared to CDK4/6i and induced tumor regression at some doses. BTX-9341 exhibited efficacy in a Palbociclib-resistant cell line and a CDK4/6 degrader showed efficacy in CDK4/6i-resistant models indicating that a degrader approach may work well in patients who are resistant to CDK4/6 inhibitors. Considering these properties, we have progressed BTX-9341 into IND enabling studies.

REFERENCES

1. Scheidemann, E.R., et al. *Int J Mol Sci* **22**, 12292 (2021).
2. Álvarez-Fernández, M., et al. *Cancer Cell* **37**, 514-529 (2020).
3. Li, Z., et al. *Cancer Cell* **34**, 893-904 (2018).
4. Li, Q., et al. *Cancer Discovery* **12**, 356-371 (2022).
5. Razavi, P., et al. *Cancer Cell* **34**, 427-438 (2018).

This presentation is the intellectual property of the author/presenter. Contact them at hmajeski@biotheryx.com for permission to reprint and/or distribute

biotheryx

http://www.biotheryx.com